ADU-1604, a novel CTLA-4 blocking antibody modulates pharmacodynamic markers in PD1 relapsed/refractory melanoma patients

P770

Guillermo de Velasco[1], Piotr Rutkowski[2], Michele Maio[3], Ana Maria Arance[4], Caroline Robert[5], Vanna Chiarion-Sileni[6], Ivan Marquez-Rodas[7], Jessica Kundapur[8], Julien Villaudy[8], Hans van Eenennaam[8], Laura Lassouw-Polman[8] and Alexander Eggermont[8,10]

1) University Hospital 12 de Octubre, Madrid, Spain; 2) Narodowy Instytut Onkologii im, Marii Skłodowskiej-Curie - Państwowy instytut Badawczy, Warszawa, Poland; 3) Università degli Studi di Siena, Siena, Italy; 4) Hospital Clínic de Barcelona, Barcelona, Spain; 5) Gustave Roussy and Paris Saclay University, Villejuif, France; 6) Veneto Oncology Institute (IOV-IRCCS), Padova, Italy; 7) Hospital General Universitario Gregorio Marañón, Madrid, Spain; 8) Sairopa b.v., Rotterdam, the Netherlands

Background

Cytotoxic T-lymphocyte-associated protein 4 (CTLA-4) is a negative regulator of T-cell responses, also known as an immune checkpoint. The clinical relevance of CTLA-4 blockade was demonstrated by the approval of ipilimumab (YERVOY®) for the treatment of melanoma, both in the adjuvant as well as metastatic settings. Sairopa acquired ADU-1604, a humanized hIgG1 CTLA-4 antagonist antibody. ADU-1604 binds a unique epitope on CTLA-4 demonstrating, in contrast to ipilimumab full blockade of both CD80 and CD86 interactions. *In vitro* and *in vivo* ADU-1604 demonstrates at least as potent efficacy as compared to ipilimumab, was well tolerated in non-human primates and demonstrated enhanced immunogenicity against hepatitis B vaccine in non-human primates[1].

(1) P335. Characterization of a novel differentiated anti-CTLA-4 antibody (ADU-1604) in vitro and in vivo. http://dx.doi.org/10.1186/s40425-017-0288-4

Methods

This is a phase 1, first-in-human (FIH), two-part, open-label clinical trial of intravenous (IV) administration of ADU-1604 given as monotherapy in subjects with advanced-stage, relapsed/refractory melanoma who relapsed or were refractory to a prior anti-PD-1/PD-L1 therapy. Main endpoints are safety of ADU-1604 monotherapy, pharmacokinetics, pharmacodynamics (upregulation of ICOS and Ki-67 on circulation CD4+ T cells and ALC, CD4+ and CD8+ T cells) as well as preliminary clinical efficacy. Up to 20 subjects will receive escalating doses of ADU-1604 IV (25, 75, 225, 450 mg flat dose) Q3W until RP2D is defined. In the dose expansion part up to 20 additional patients will be treated at the recommended phase 2 dose (RP2D) to further characterize safety, PK, PD and explore initial clinical activity of ADU-1604 monotherapy in PD1 relapsed/refractory melanoma patients.
The study was initiated in June 2022. Cohort 4 (450 mg) is ongoing at time of submission of the abstract (07/2023). The clinical trial is registered with EudraCT number 2021-002623-38.

ADU-1604 binds unique epitope on CTLA-4

ADU-1604 PDB ID: n.a. mapped on 5GGV | **ipilimumab** PDB ID: 5TRU mapped on 5GGV | **tremelimumab** mapped on 5GGV | **Gotistobart** PDB ID: 6XY2 mapped on 5GGV

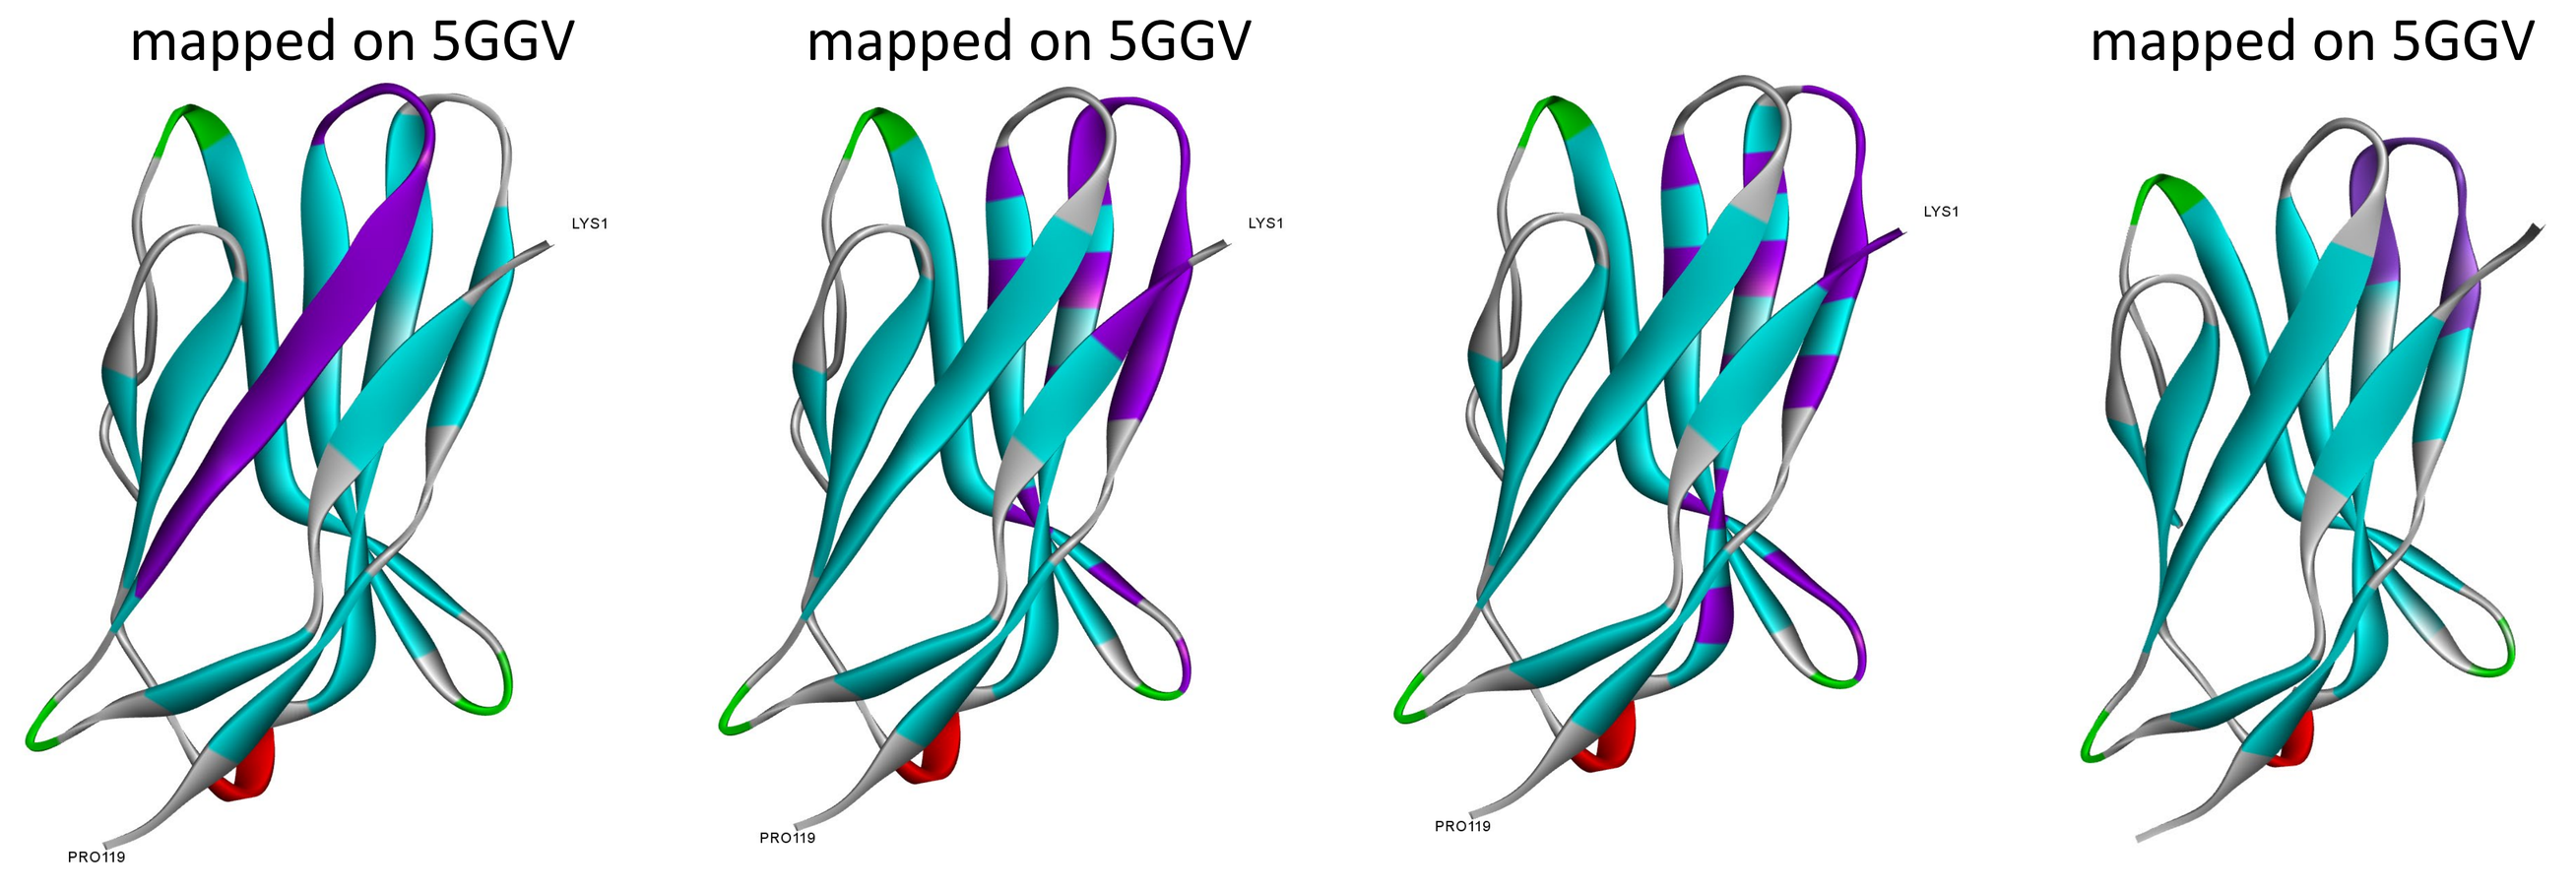

The epitope of ADU-1604 was determined by deuterated chemical crosslinking followed by enzymatic digestion and peptide mass fingerprinting using mass spectrometry (CovalX, Zurich, Switzerland) and highlighted in purple. The same experimental procedure was used to determine the binding region of ipilimumab. The epitope residues determined for ipilimumab overlap with the residues identified in the X-ray structure as described by Ramagopal et al (2017) PNAS 114: E4223-4232 (PDB ID: 5TRU). The tremelimumab and Gotistobart binding region was not determined here, its graphic representation is based on the epitope as described by Lee et al (2016) Nat Commun 7: 13354-13354 and Gao et al. Cell Discovery (2020)6:79, respectively mapped on PDB ID: 5GGV.

ADU-1604 SHOWS pH-SENSITIVE BINDING TO CTLA-4

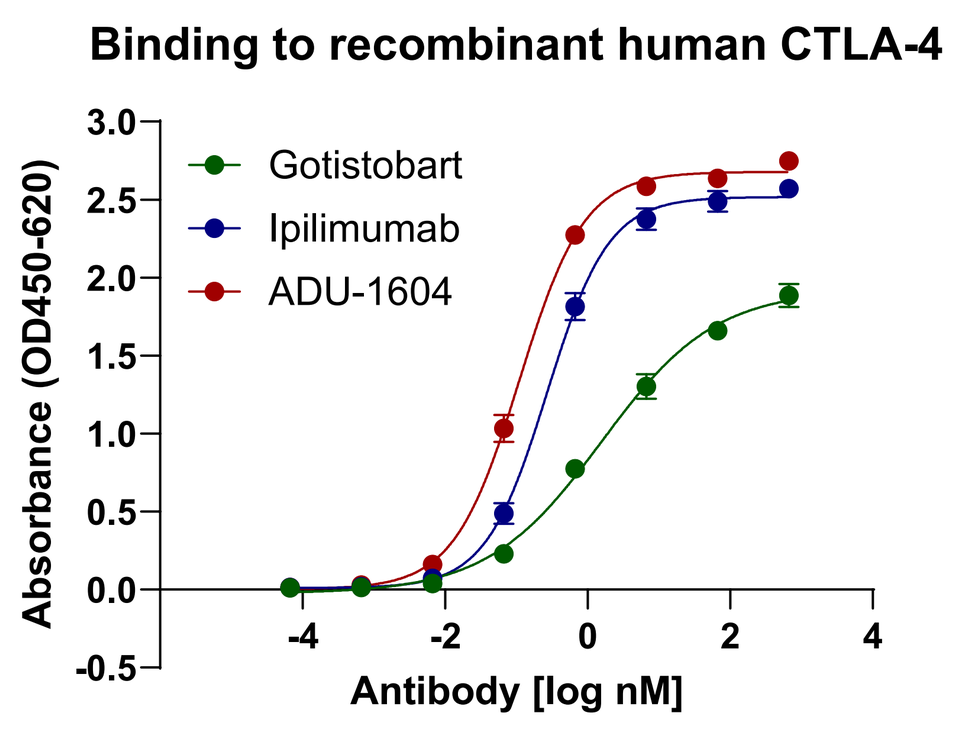
Binding to recombinant human CTLA-4
Gotistobart
Ipilimumab
ADU-1604
Absorbance (OD450-620)
Antibody [log nM]

	Gotistobart	Ipilimumab	**ADU-1604**
Top	**1.923**	2.517	2.678
EC50	1.573	0.2709	**0.1102**

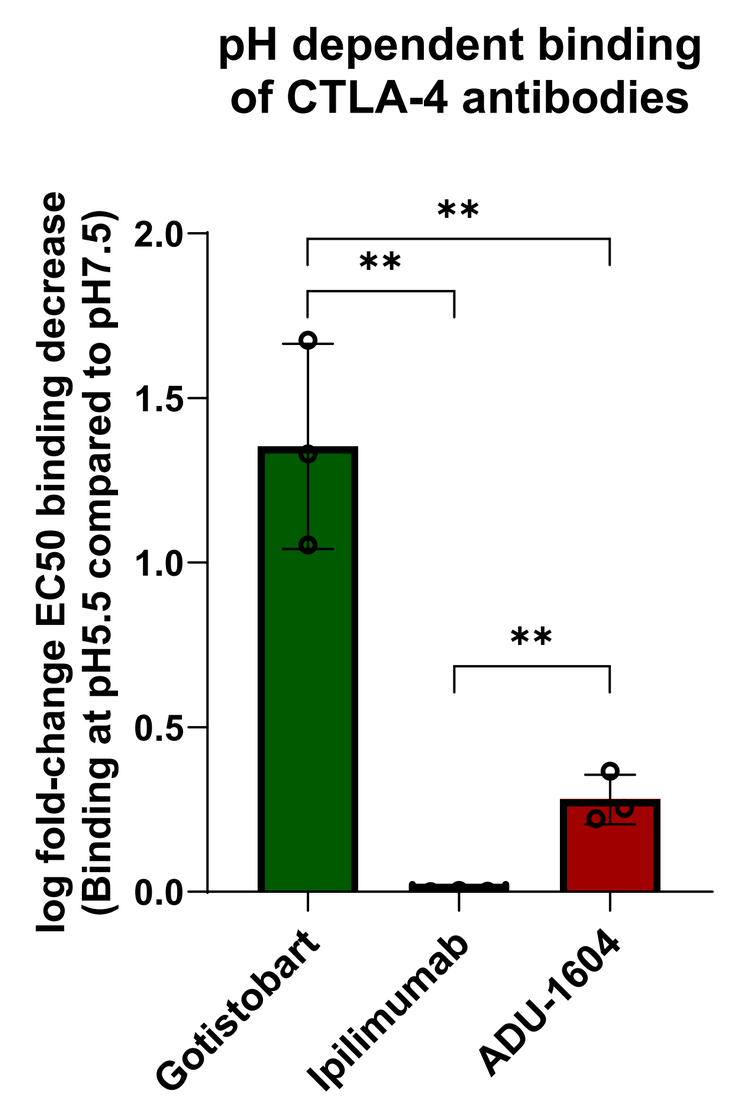
pH dependent binding of CTLA-4 antibodies
log fold-change EC50 binding decrease (Binding at pH5.5 compared to pH7.5)
Gotistobart
Ipilimumab
ADU-1604

ADU-1604 demonstrates strongest binding potency to human recombinant CTLA-4 as compared to ipilimumab and Gotistobart. Of note, ADU-1604 and ipilimumab demonstrate a higher binding efficacy as compared to Gotistobart (left panel). Comparing binding of ADU-1604, ipilimumab and Gotistobart at pH of 7.5 vs. 5.5 (late endosome), ADU-1604 and Gotistobart demonstrate pH-sensitivity for binding to recombinant CTLA-4 when compared to ipilimumab.

SRP-21C101: PHASE 1 DOSE ESCALATION AND EXPANSION

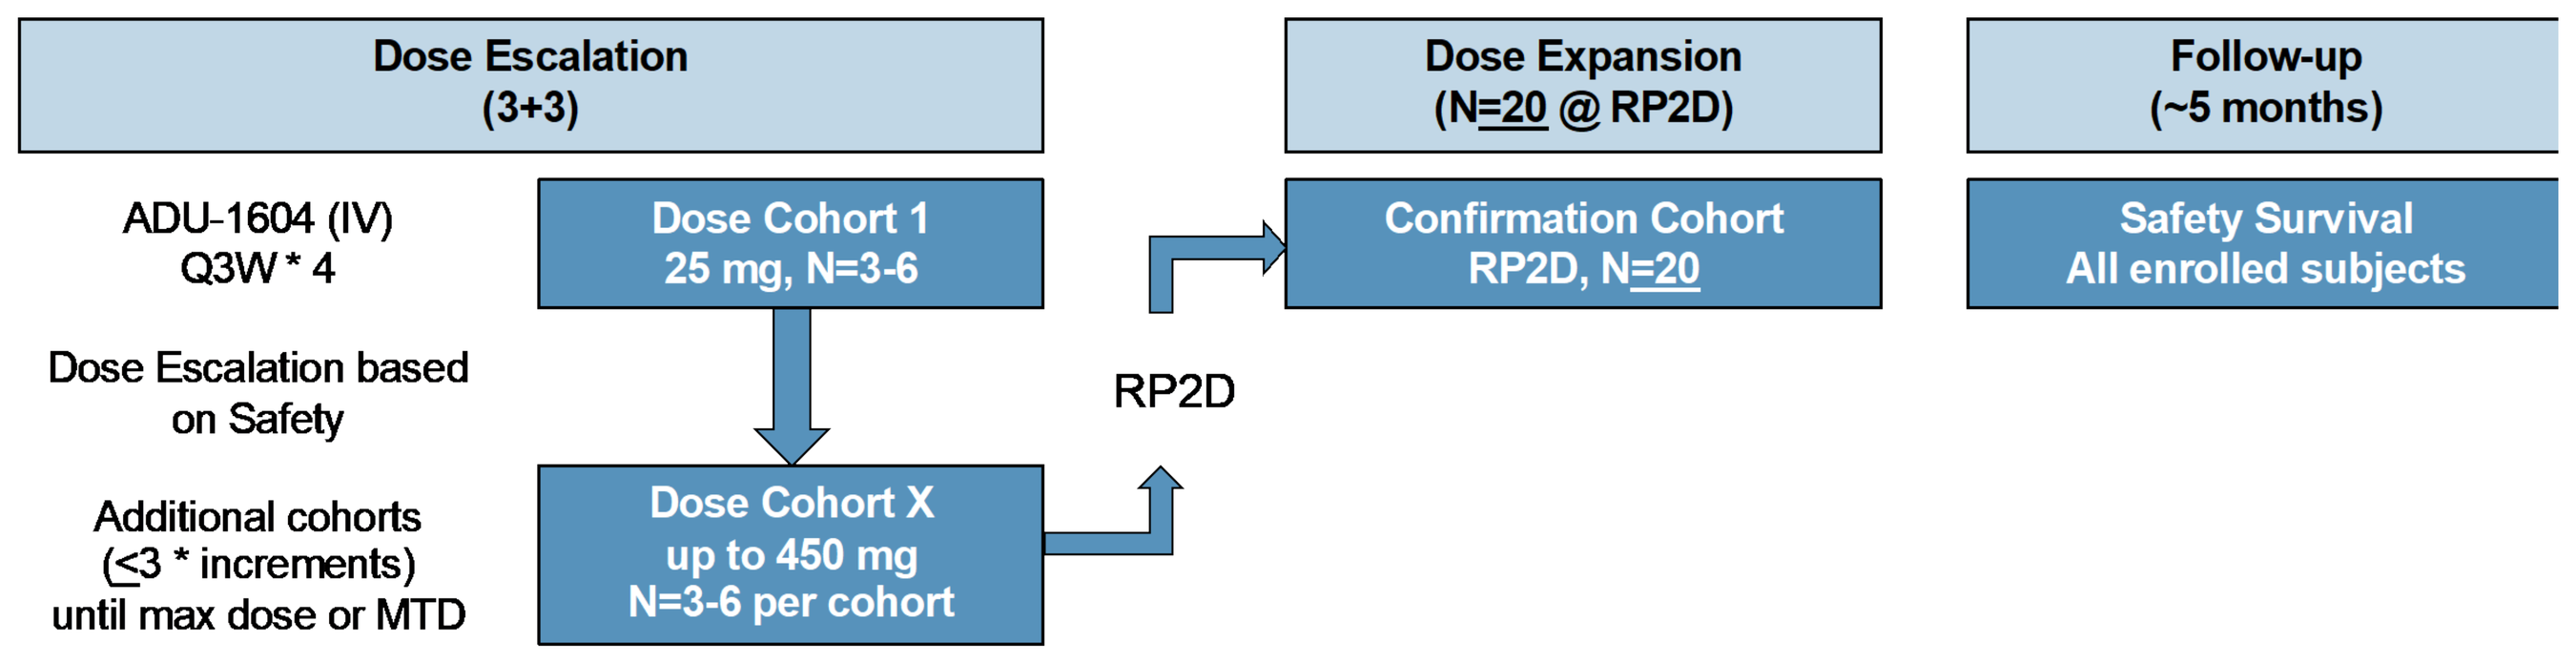
Dose Escalation (3+3)
ADU-1604 (IV) Q3W * 4
Dose Escalation based on Safety
Additional cohorts (≤3 * increments) until max dose or MTD
Dose Cohort 1 25 mg, N=3-6
Dose Cohort X up to 450 mg N=3-6 per cohort
RP2D
Dose Expansion (N=20 @ RP2D)
Confirmation Cohort RP2D, N=20
Follow-up (~5 months)
Safety Survival All enrolled subjects

PATIENT CHARACTERISTICS

	25 mg* Q3W (N = 4)	75 mg Q3W (N = 4)	225 mg Q3W (N = 6)	450 mg Q3W (N = 3)	Total (N = 17)
Age (years)					
Mean	63.3	61.8	67.6	51.3	63.7
Sex (N (%))					
Male	2 (50)	4 (100)	5 (83)	3 (100)	14 (83)
Female	2 (50)	0 (0)	1 (17)	0 (0)	3 (17)
Melanoma type					
Nodular Melanoma	2 (50)	1 (25)	2 (33,3)	2 (67)	7 (41)
Superficial Spreading Melanoma	1 (25)	0 (0)	2 (33,3)	1 (33)	4 (23,5)
Lentigo Maligna	1 (25)	3 (75)	0 (0)	0 (0)	4 (23,5)
Other	0 (0)	0 (0)	2*(33,3)	0 (0)	2 (12)
ECOG Performance Status					
0	2	2	4	2	10
1	2	2	2	1	7
Prior treatment line(s) (n[%])					
1-2	4 (100)	4 (100)	5 (83)	1 (33)	14 (83)
3-4	0 (0)	0 (0)	1 (17)	2 (67)	3 (17)
PD1/PDL1 inhibitors	4 (100)	4 (100)	6 (100)	3 (100)	17 (100)
Number of Cycles of ADU-1604 (1 Cycle ~ 21 days)					
Average	8	2	4	2	4

* One patient received 25 mg for cycles 1-8, then received 75mg from Cycle 9-13
** Other subtypes were Epithelioid Cell Melanoma and Acral Lentiginous Melanoma
Database cut-off: 28 September 2023

450 MG DOSE OF ADU-1604 IS WELL TOLERATED

All data are n (%)	25 mg Q3W (N = 4)		75 mg Q3W (N = 4)		225 mg Q3W (N = 6)		450 mg Q3W (N = 3)		Total (N = 17)	
	Grade 1-2	Grade 3-4*	Grade 1-2	Grade 3	Grade 1-2	Grade 3	Grade 1-2	Grade 3	Grade 1-2	Grade 3-4*
Adverse Events (AEs) of any cause	4 (100)	3 (50)	4 (100)	1 (25)	6 (100)	3 (50)	3 (100)	0 (0)	17 (100)	7 (41)
AEs leading to treatment discontinuation	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)
Serious Adverse Event (SAE)	-	0 (0)	-	0 (0)	-	2** (33)	-	0 (0)	0 (0)	2 (12)
Adverse Events of Special Interest (AESI)	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)	2*** (33)	0 (0)	0 (0)	0 (0)	2 (12)
Treatment-related AEs****	3 (75)	0 (0)	0 (0)	0 (0)	5 (83)	2 (33)	3 (100)	0 (0)	10 (59)	2 (12)

*One Grade 4 event; Neutropenia unrelated to ADU-1604. No grade 5 events occurred.
**SAE was a femur fracture, unrelated to ADU-1604 & Grade 3 fever requiring hospitalization, unrelated to ADU-1604
*** Two AESIs; Grade 3 Immune-mediated gastritis, probably related to ADU-1064 & Grade 3 Alanine aminotransferase increased, possibly related to ADU-1604.
**** Most common treatment-related AEs were Rash [2/17 (12%)] & Pruritis [2/17 (12%)] reported in the 225mg cohort and Fatigue [2/17 (12%)] reported in the 450mg cohort.
Database cut-off: 28 September 2023

ADU-1604 DEMONSTRATES EARLY SIGNS OF CLINICAL EFFICACY

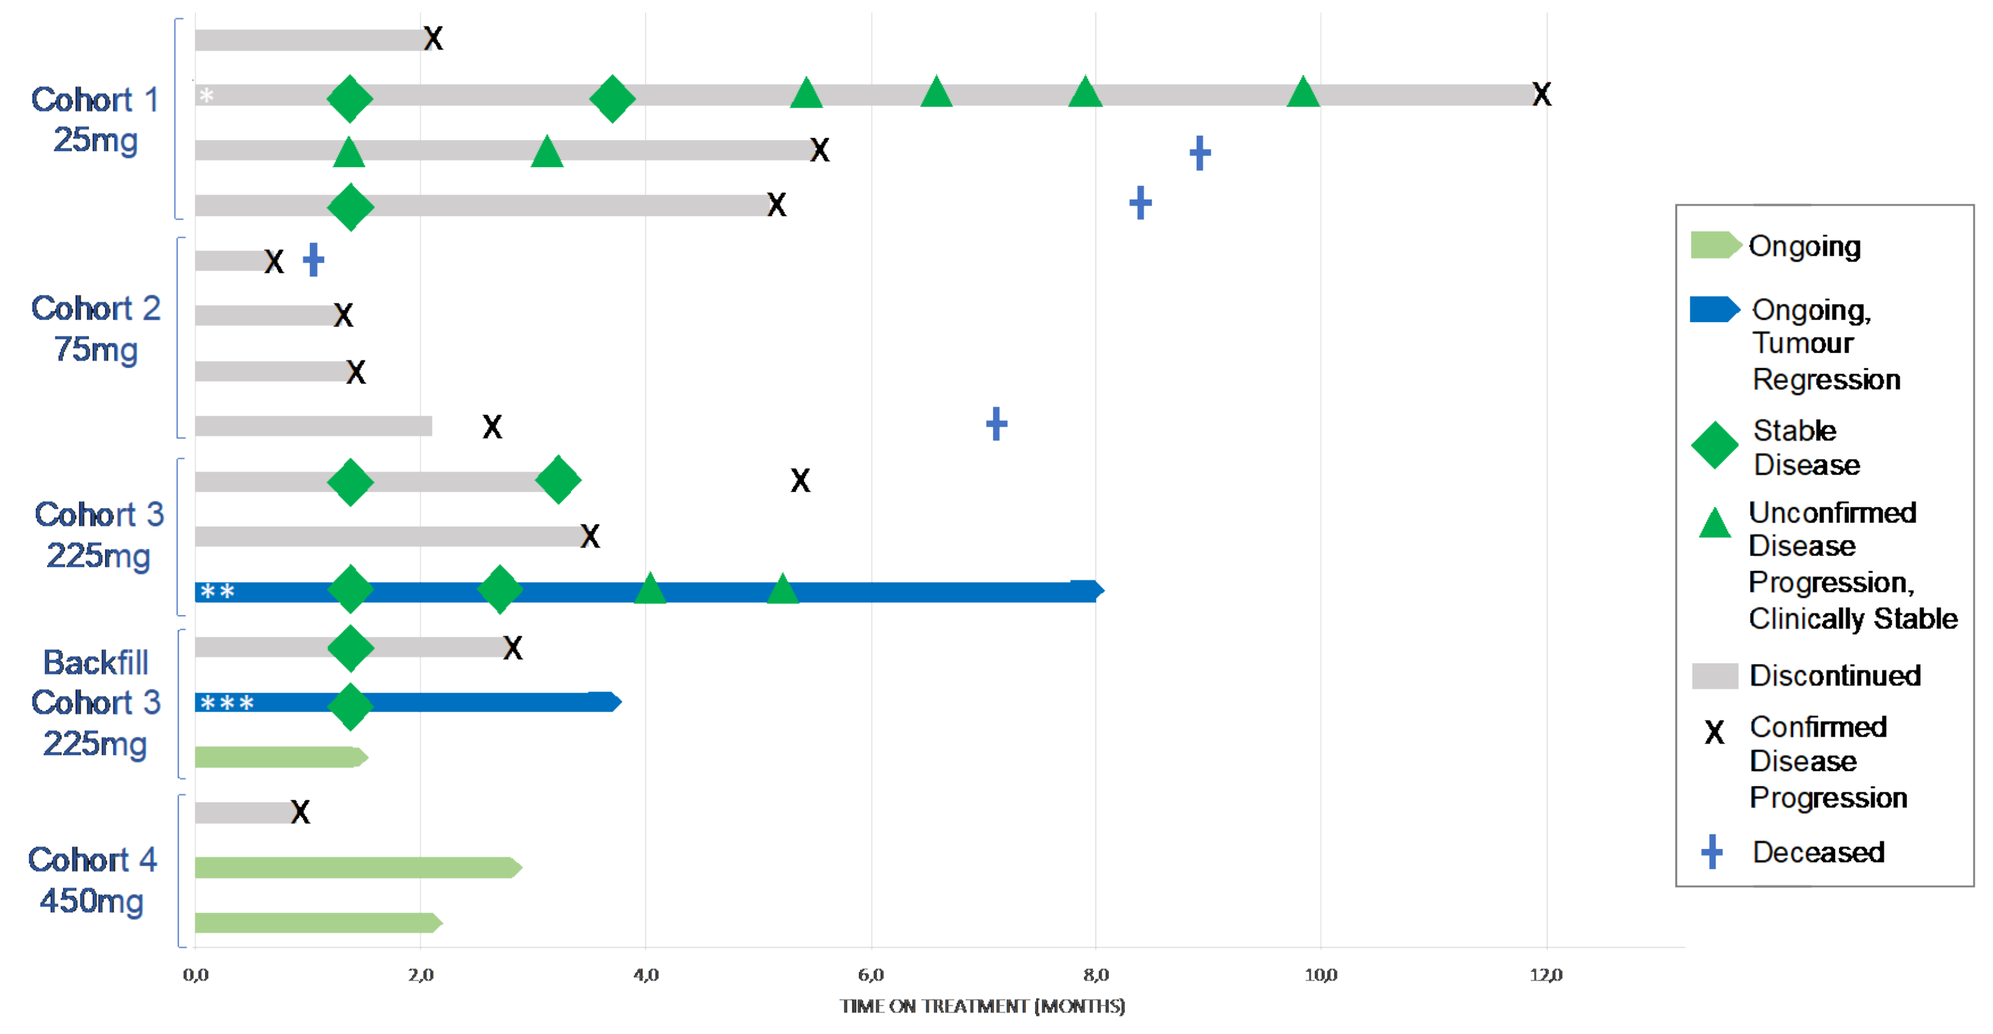
Cohort 1 25mg
Cohort 2 75mg
Cohort 3 225mg
Backfill Cohort 3 225mg
Cohort 4 450mg
Ongoing
Ongoing, Tumour Regression
Stable Disease
Unconfirmed Disease Progression, Clinically Stable
Discontinued
Confirmed Disease Progression
Deceased
TIME ON TREATMENT (MONTHS)

* Patient received 25 mg from Cycle 1-Cycle 8, received 75mg from Cycle 9 to Cycle 13. Confirmed disease progression at 11,9 months.
** Patient has tumour decrease of -20.5%
*** Patient has tumour decrease of -26.5%
Database cut-off: 28 September 2023

ADU-1604 DOSE-DEPENDENTLY INCREASES KI-67 POSITIVE T CELLS

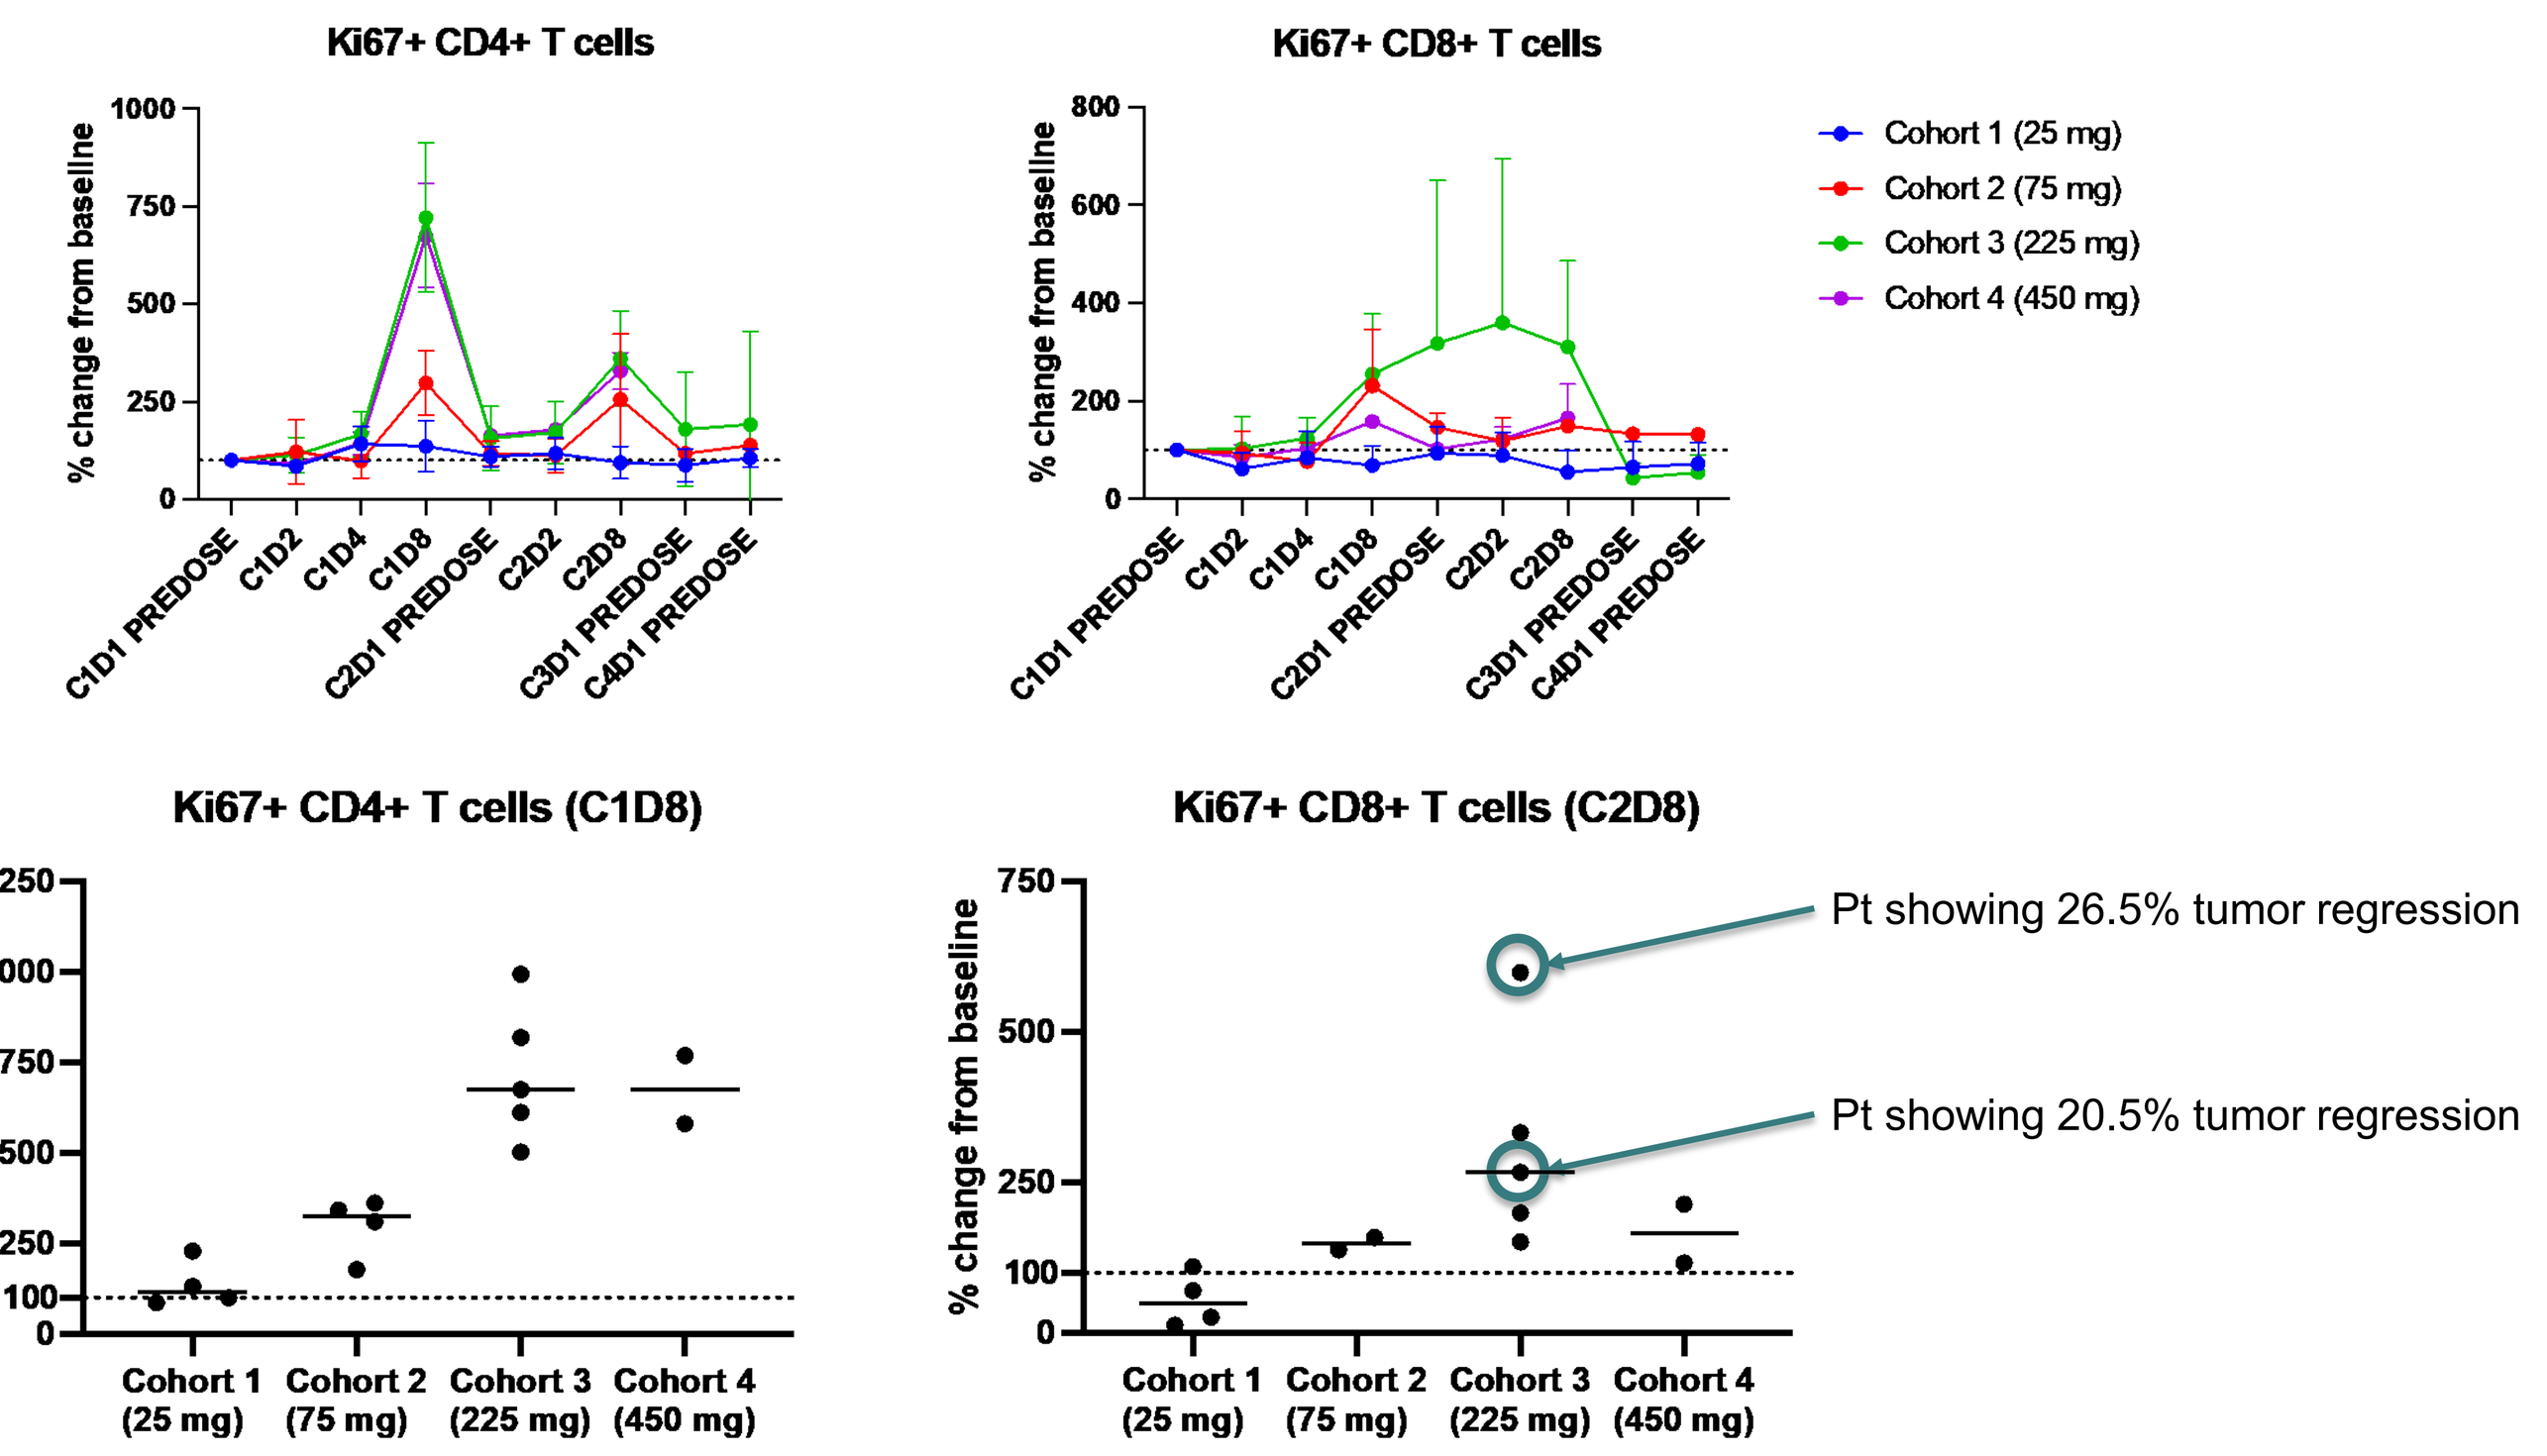
Ki67+ CD4+ T cells
Ki67+ CD8+ T cells
Cohort 1 (25 mg)
Cohort 2 (75 mg)
Cohort 3 (225 mg)
Cohort 4 (450 mg)
% change from baseline
C1D1 PREDOSE
C1D2
C1D4
C1D8
C2D1 PREDOSE
C2D2
C2D8
C3D1 PREDOSE
C4D1 PREDOSE
Ki67+ CD4+ T cells (C1D8)
Ki67+ CD8+ T cells (C2D8)
Cohort 1 (25 mg)
Cohort 2 (75 mg)
Cohort 3 (225 mg)
Cohort 4 (450 mg)
Pt showing 26.5% tumor regression
Pt showing 20.5% tumor regression

ADU-1604 is dose-dependently inducing Ki-67 (proliferation marker) after the first and second administration, indication that ADU-1604 enhances T cell proliferation. Increased proliferation of CD4 and CD8 T cells is observed upon ADU-1604 administration (top panel, left and right respectively). The bottom panels present the enhanced Ki-67 positive CD4 and CD8 T cells in individual patients dosed with 25, 75, 225 and 450 mg on C1D8 and C2D8, respectively.

Summary

✓ ADU-1604 is a novel CTLA-4 blocking antibody that targets a unique epitope on CTLA-4.
✓ ADU-1604 binds CTLA-4 strongest as compared to other CTLA-4 targeting antibodies and demonstrates pH sensitive binding to CTLA-4.
✓ In melanoma patients, relapse/refractory to PD-1, blockade ADU-1604 is well tolerated up to 450 mg dose.
✓ Induction of proliferation of CD4 and CD8 T cells is induced by ADU-1604, coinciding with early signs of clinical activity.

Ethics Approval: All participants gave informed consent prior to study participation. Ethical Committee Approval issued by France, CPP Sud-Méditerranée on16 Mar 2022, issued by Spain, CEim Hospital Clinic on 16 Mar 2022, issued by Italy, IRCCS Pascale on 31 Mar 2022, issued by Poland, Bioethics Committee of Narodowy Instytut Onkologii on 15 Dec 2021.

SCAN QR CODE
www.Sairopa.com